Poster 644

Prophylactic administration of the monoclonal antibody ADG20 provides potent protection against SARS-CoV-2 in rodent and non-human primate models of COVID-19

Kristin Narayan,[1] Chengzi I. Kaku,[1,2,*] Laura M. Walker,[1,2] Lynn Connolly,[1] Ellie Hershberger,[1] Ilker Yalcin,[1] Zoltán Magyarics,[1] Andrew S. Herbert,[3] Elizabeth E. Zumbrun,[3] Cecilia M. O'Brien,[3] Russell R. Bakken,[3] Samantha E. Zak,[3] John M. Dye[3]

[1]Adagio Therapeutics, Inc., Waltham, MA, USA; [2]Adimab LLC, Lebanon, NH, USA; [3]US Army Medical Research Institute of Infectious Diseases, Frederick, MD, USA

*Presenting author

INTRODUCTION

- The spread of severe acute respiratory syndrome coronavirus 2 (SARS-CoV-2), which causes coronavirus disease (COVID-19), and the emergence of new variants of concern resistant to some antibody-based therapeutics, is an ongoing global public health issue; the search for therapies to treat or prevent this disease remains a matter of urgency[1,2]
- ADG20 is a highly potent, broadly neutralising fully human immunoglobulin (IgG)1 monoclonal antibody (mAb) derived from a survivor of the 2003 SARS-CoV epidemic and modified to have an extended half-life[3]
- ADG20 targets a highly conserved epitope on the receptor binding domain (RBD) of the spike protein of SARS-CoV-2, SARS-CoV, and other pre-emergent SARS-like CoVs[3]
- In vitro ADG20 displays high binding affinity and potent neutralisation against all SARS-CoV-2 variants tested, including variants of concern (B.1.1.7/Alpha, B.1.351/Beta, P.1/Gamma)[3,4]
- Preclinical studies have demonstrated prophylactic protection against mouse-adapted SARS-CoV and SARS-CoV-2[3]
- Here we report two studies that evaluated the in vivo prophylactic efficacy of ADG20 in Syrian hamster and non-human primate (NHP) models, which were infected with actual SARS-CoV-2 virus, to investigate the impact of ADG20 on the diverse range of COVID-19 manifestations
 - Additionally, we evaluated the risk for antibody-dependent enhancement (ADE) at subneutralising doses of ADG20 in hamsters

METHODS

Syrian hamster model (Figure 1A)

- Syrian hamsters were dosed intraperitoneally (IP) with either ADG20 (n=40; 9.25–2000 µg) or sham-treated isotype-matched IgG (n=20; either 9.25 or 2000 µg) 24 hours prior to intranasal (IN) SARS-2/WA1 challenge (1×10^5 plaque-forming units [PFU])
- Hamsters were weighed daily over 6 days, while antibody titres, viral load, and lung histopathology were assessed following sacrifice of five animals from each group on Days 3 and 6 post-challenge

NHP model (Figure 1B)

- Rhesus macaques were dosed intravenously (IV) with either ADG20 (n=8; 5 or 25 mg/kg) or sham-treated isotype-matched IgG (n=4; 25 mg/kg) 72 hours prior to IN/intratracheal (IT) SARS-2/WA1 challenge (1×10^6 PFU)
- Viral load was assessed in nasopharyngeal (NP) and oropharyngeal (OP) samples (daily) and bronchoalveolar lavage (BAL; Days 1, 3, and 5) by reverse transcription-polymerase chain reaction (RT-PCR; all samples) and plaque assay (NP and OP swabs only)

Figure 1. Study design

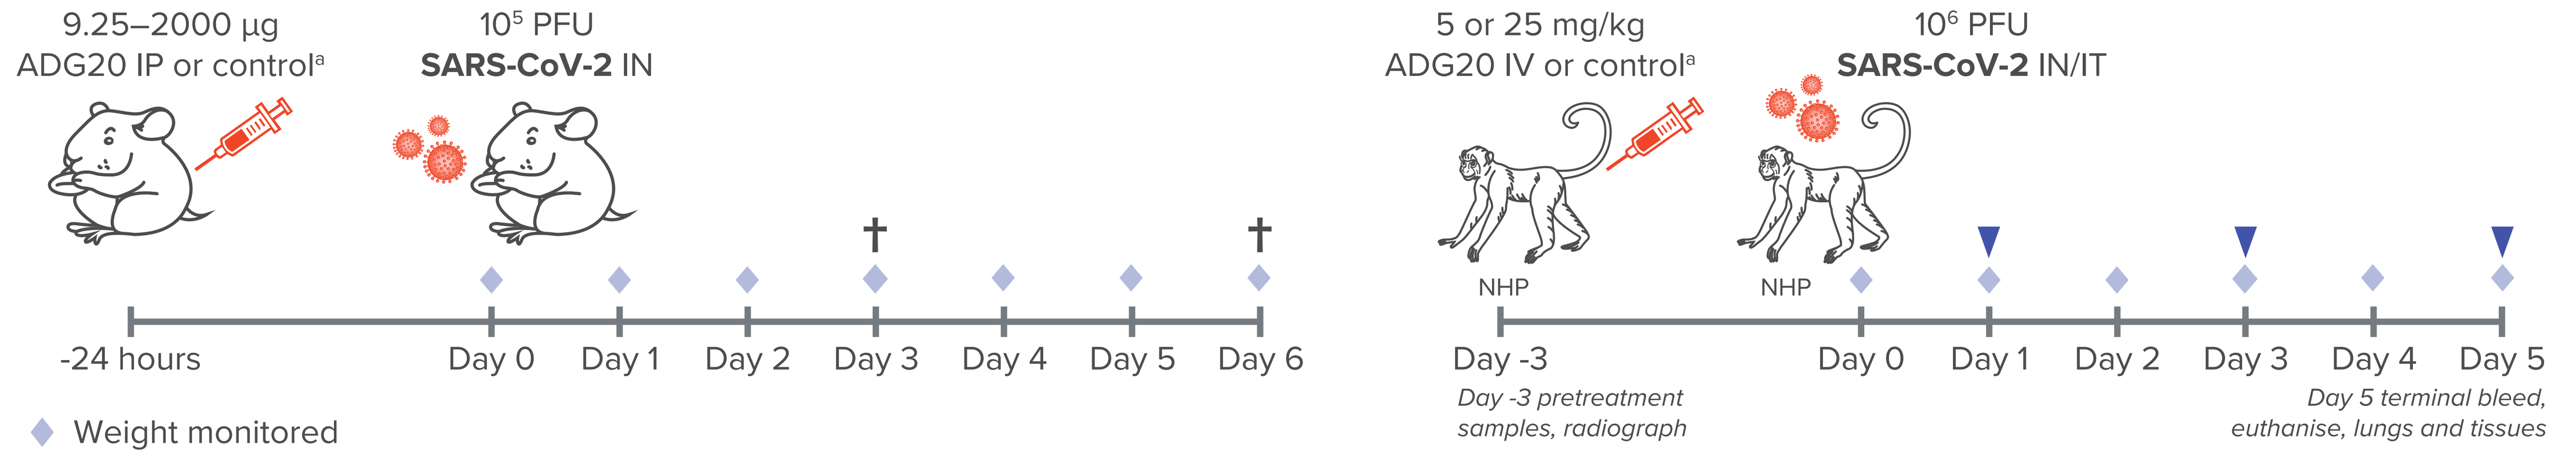
A. Syrian hamster model
9.25–2000 µg
ADG20 IP or control[a]
10^5 PFU
SARS-CoV-2 IN
-24 hours
Day 0
Day 1
Day 2
Day 3
Day 4
Day 5
Day 6
Weight monitored
Hamsters euthanised for lung viral load and histopathology analysis
B. NHP Model
5 or 25 mg/kg
ADG20 IV or control[a]
10^6 PFU
SARS-CoV-2 IN/IT
NHP
NHP
Day -3
Day -3 pretreatment samples, radiograph
Day 0
Day 1
Day 2
Day 3
Day 4
Day 5
Day 5 terminal bleed, euthanise, lungs and tissues
NP and OP viral load samples
BAL viral load samples

[a]Sham-treated isotype matched IgG.

- Note: 20 animals (n=7, control[a]; n=13, 9.25–2000 µg ADG20) were excluded from analysis due to undetectable human IgG titres after IP dosing

RESULTS

Prophylactic efficacy of ADG20 in Syrian hamsters

- An ADG20 dose of ≥55 µg was associated with protection from weight loss compared with controls at Day 6 (**Figure 2A**)
- Hamsters receiving 333 and 2000 µg doses displayed limited histopathological evidence of pneumonia (**Figure 2B**)
- Hamsters receiving the highest dose (2000 µg) had no detectable virus in lung samples (**Figure 3**):
 - 333 µg and 2000 µg ADG20 treatment significantly reduced infectious viral loads at Day 3 post-exposure compared with controls (P <0.005)
 - Similar trends were observed for viral sub-genomic RNA (sgRNA), which is produced during active viral infection
- No evidence of ADE was observed across ADG20 doses, including subtherapeutic doses

Figure 2. Impact of ADG20 on weight gain and lung pathology in SARS-CoV-2–infected Syrian hamsters

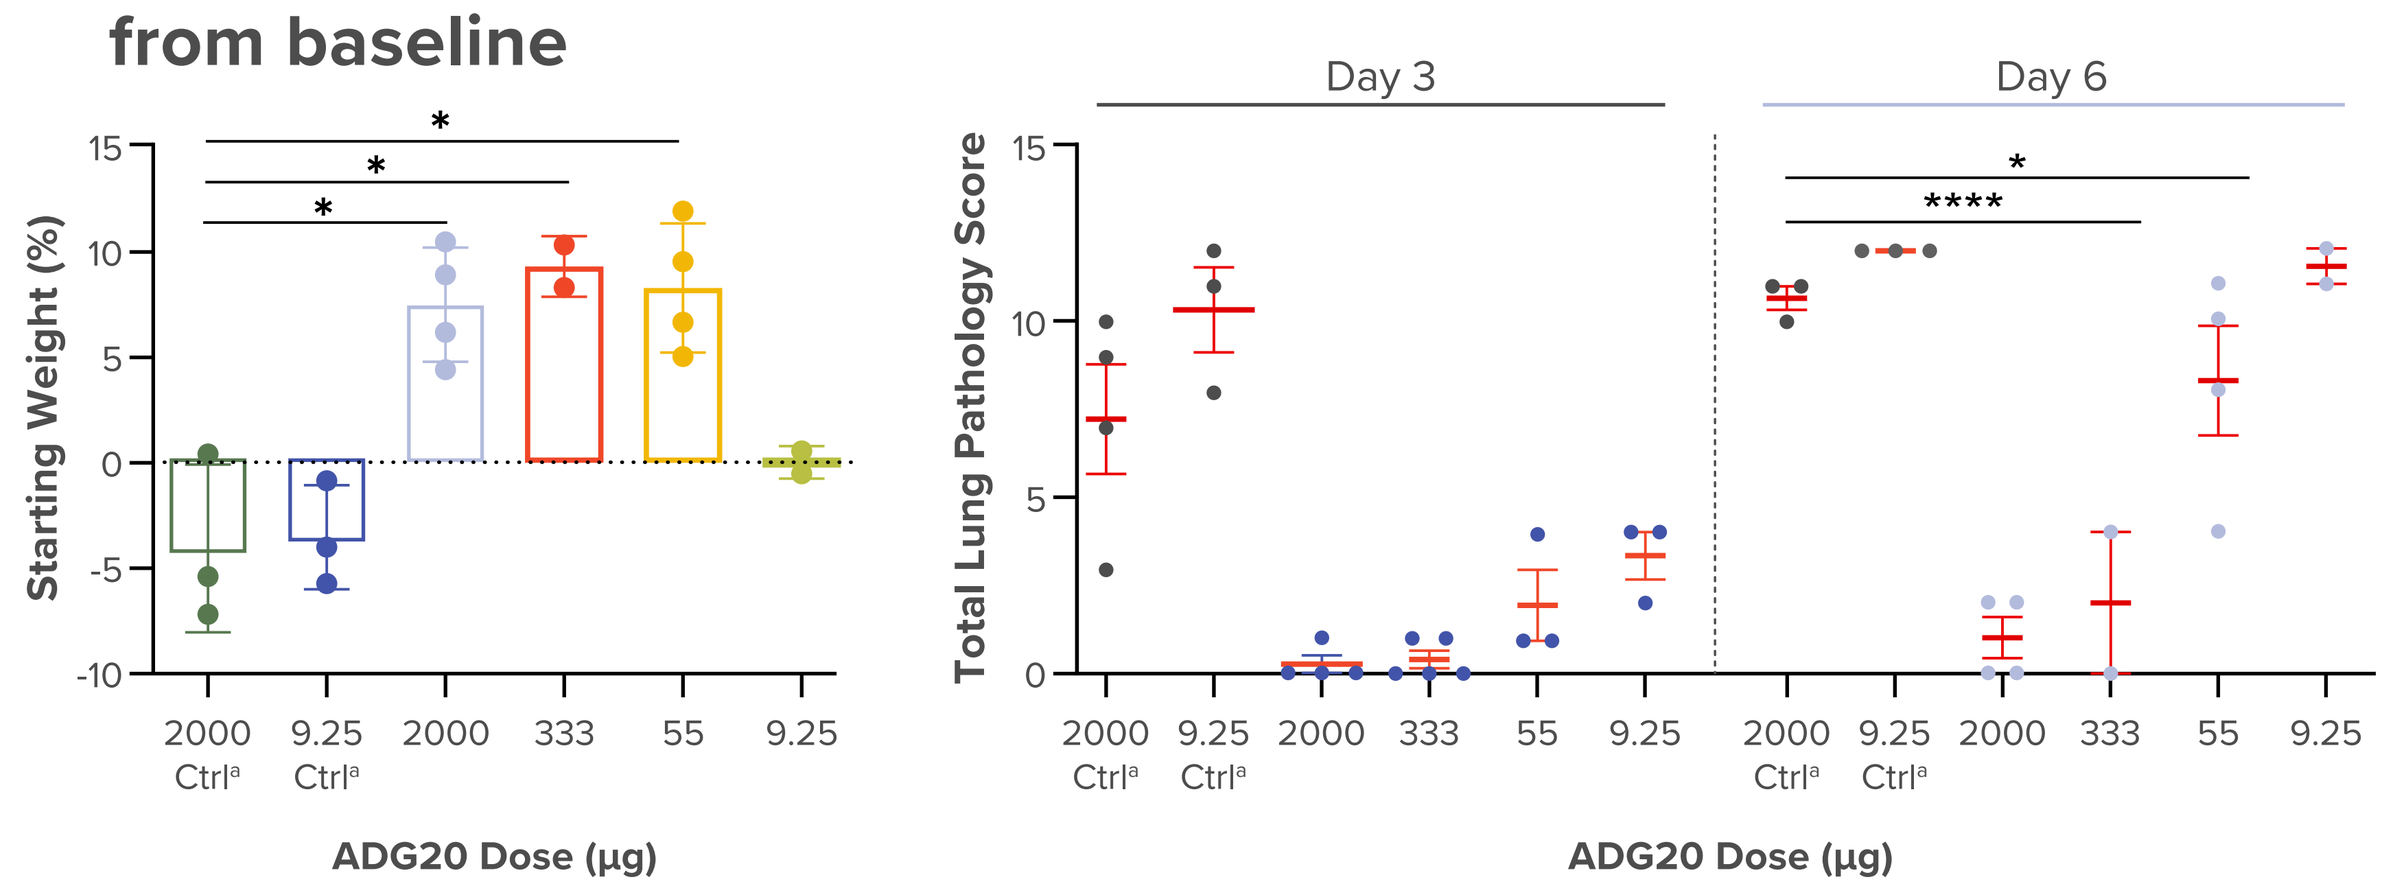
A. Day 6 weight change from baseline
B. Total lung pathology score
Starting Weight (%)
Total Lung Pathology Score
Day 3
Day 6
2000 Ctrl[a]
9.25 Ctrl[a]
2000
333
55
9.25
ADG20 Dose (µg)

Ctrl, control.
Red bars represent mean ± standard deviation. Dotted line represents no change in weight from baseline.
[a]Sham-treated isotype matched IgG.
Statistical comparison was calculated using unpaired, two-sided t-tests (A) and unpaired two-sided Mann-Whitney tests (B): *P <0.05; ****P <0.0001 vs Ctrls.

Figure 3. Impact of ADG20 on viral load in SARS-CoV-2–infected Syrian hamsters

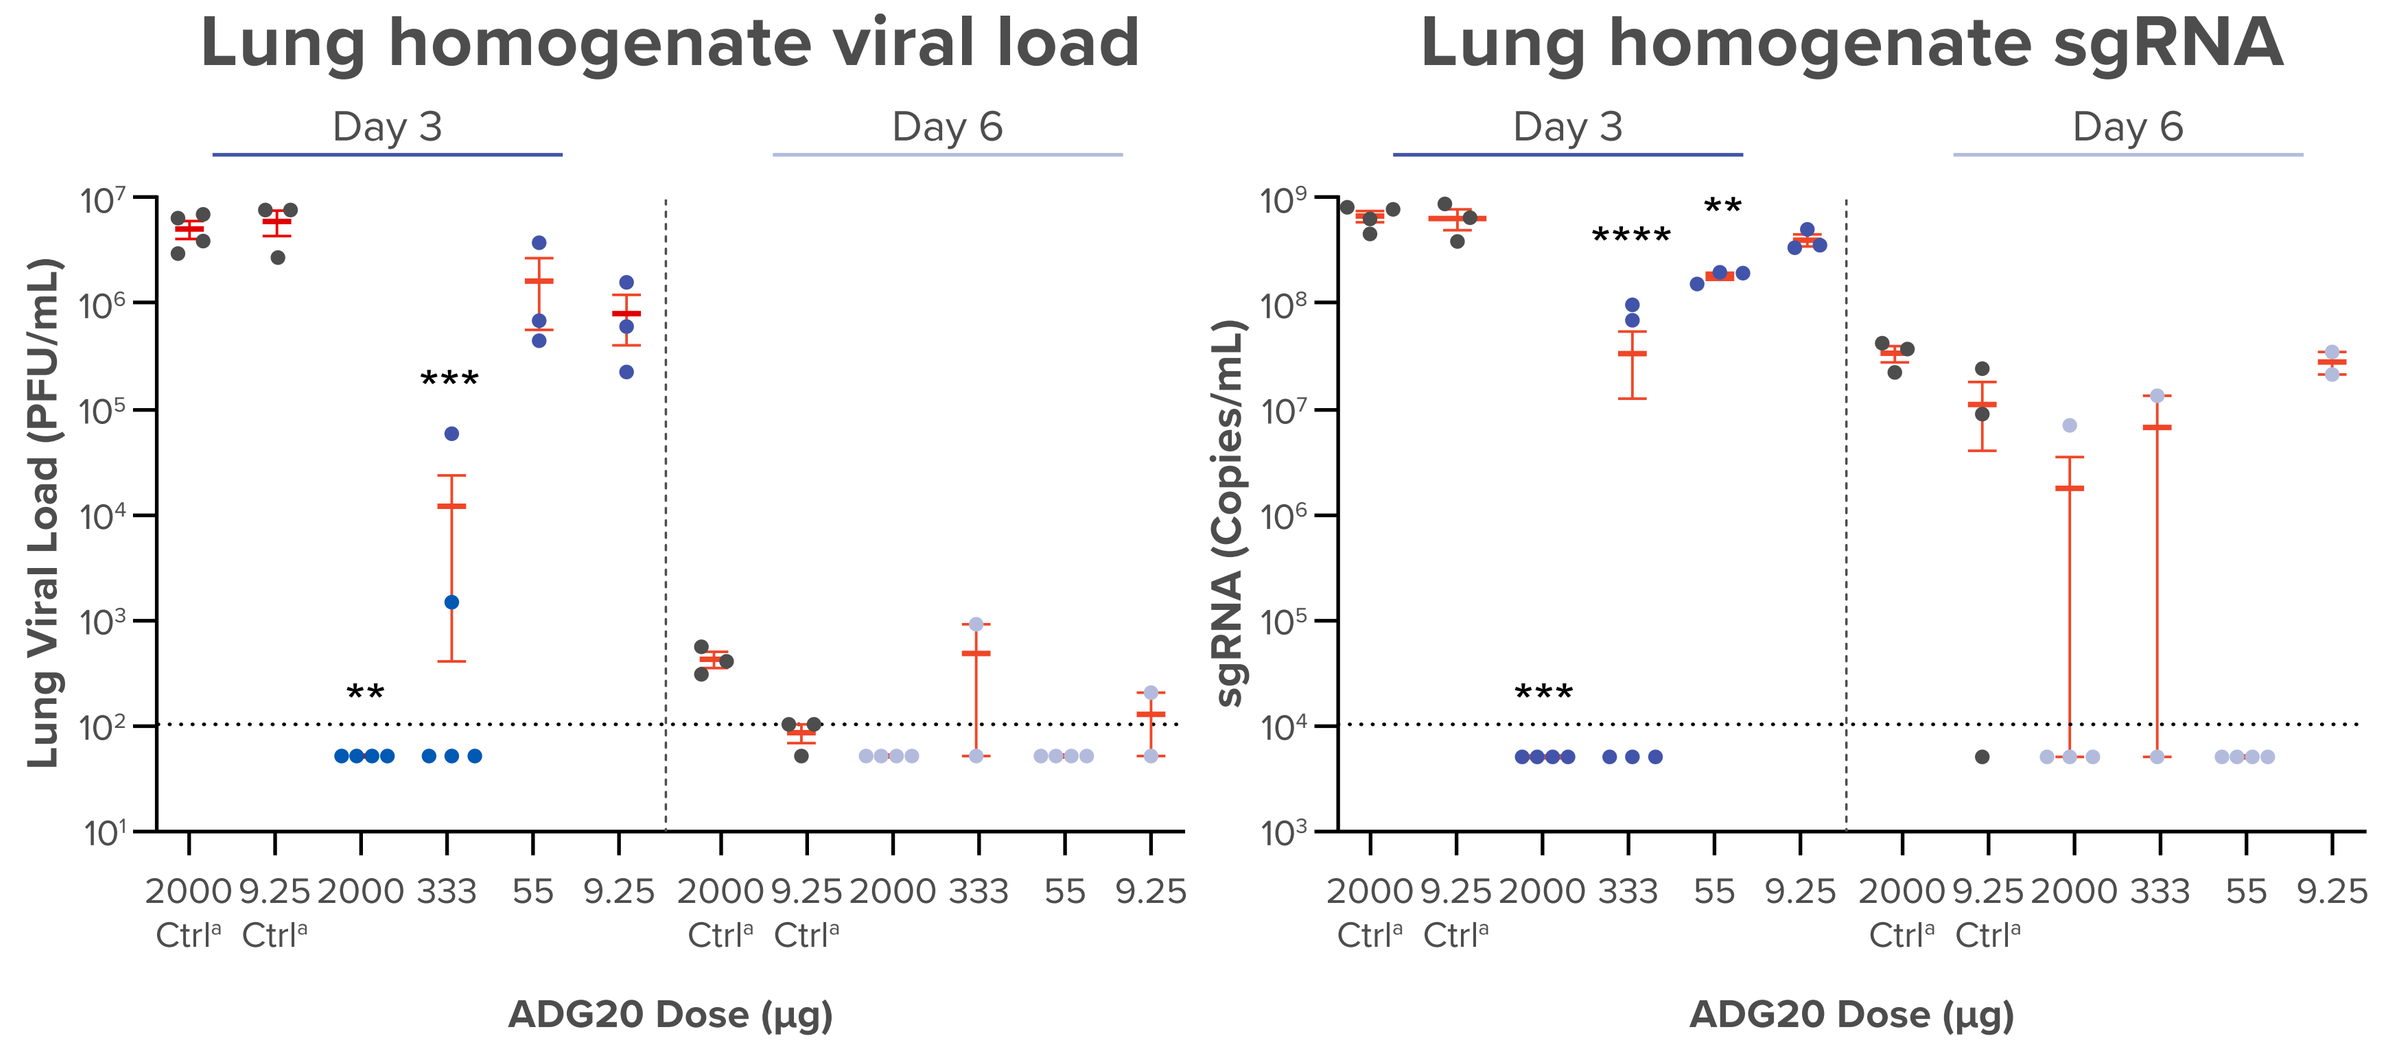
Lung homogenate viral load
Lung homogenate sgRNA
Day 3
Day 6
Lung Viral Load (PFU/mL)
sgRNA (Copies/mL)
2000 Ctrl[a]
9.25 Ctrl[a]
2000
333
55
9.25
ADG20 Dose (µg)

Red bars represent mean ± standard deviation. Dotted line represents limit of detection.
[a]Sham-treated isotype-matched IgG.
Statistical comparison was calculated using unpaired, two-sided t-tests: **P<0.005; ***P<0.001; ****P<0.0001 vs Ctrls.

Prophylactic efficacy of ADG20 in Rhesus macaques

- An ADG20 dose of 25 mg/kg was associated with reduced viral replication in the upper and lower airways with sgRNA below the limit of detection in all respiratory compartments tested (**Figure 4**):
 - A similar trend was observed for genomic RNA (data not shown)
 - Substantial protection was also observed at the 5 mg/kg dose level, as demonstrated by accelerated clearance of infectious virus
- No viral particles were detected in NP and OP compartments at the 25 mg/kg ADG20 dose (**Figure 5**)

Figure 4. Impact of ADG20 on viral replication in Rhesus macaques

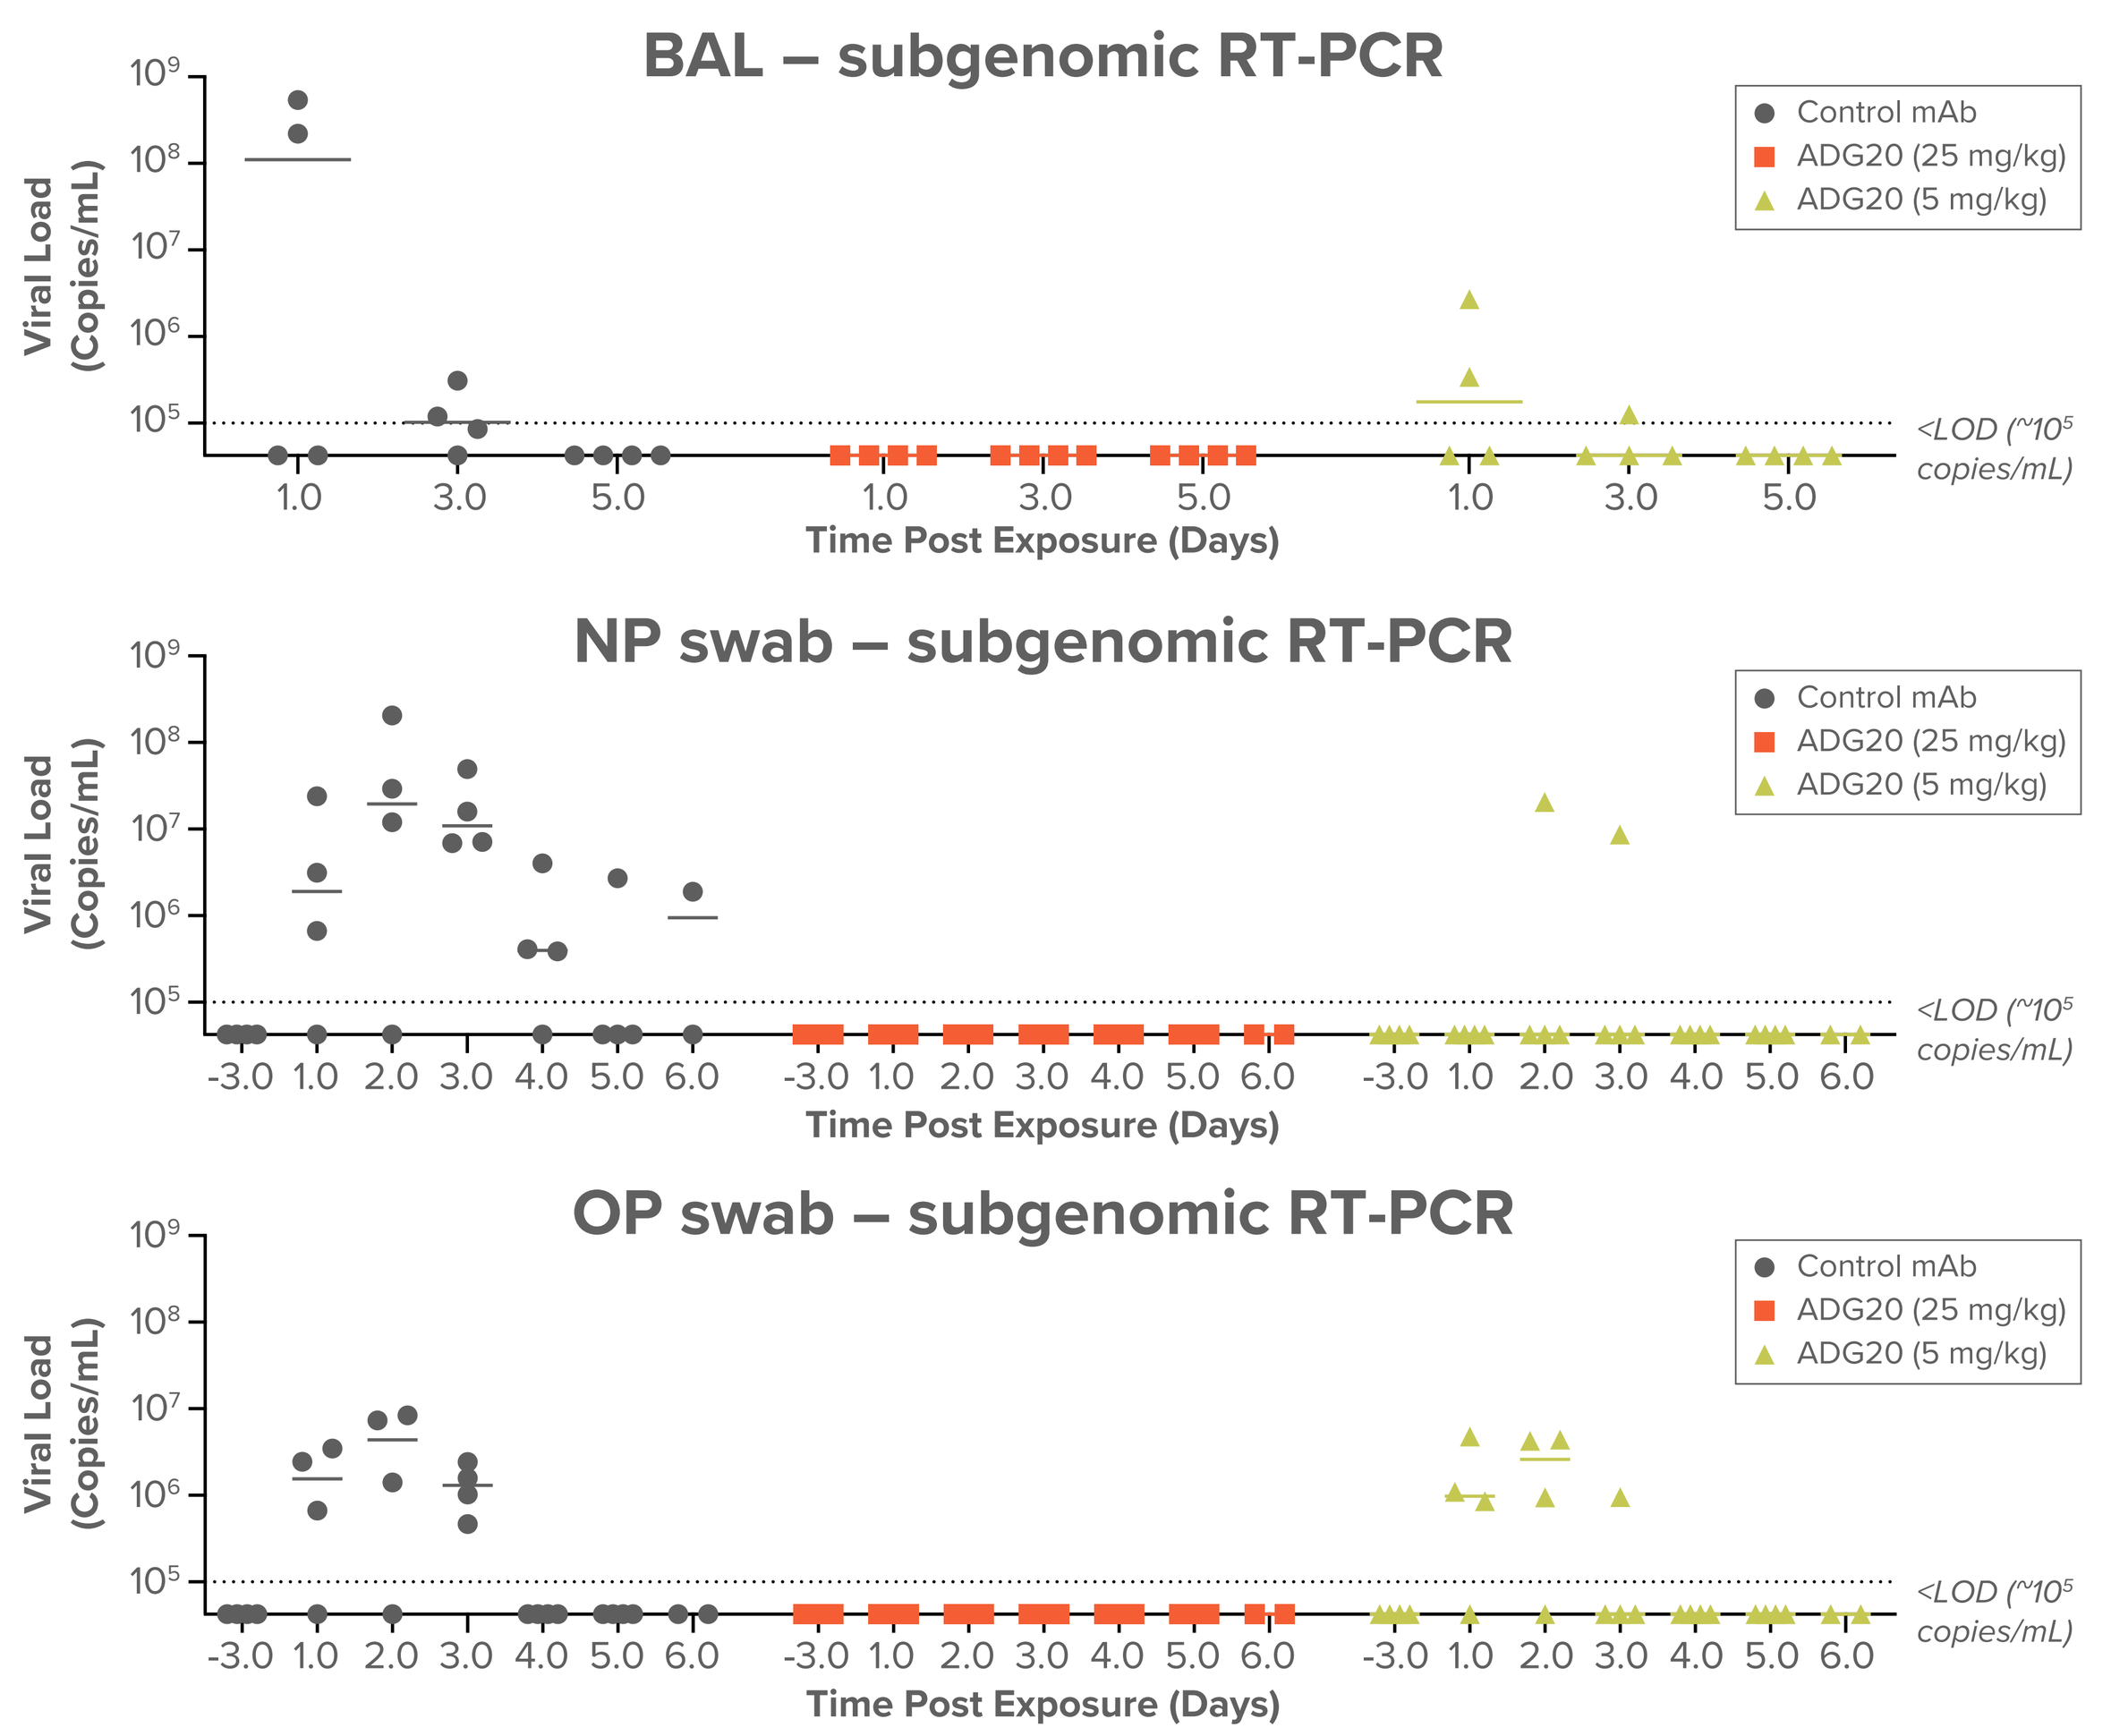
BAL – subgenomic RT-PCR
NP swab – subgenomic RT-PCR
OP swab – subgenomic RT-PCR
Viral Load (Copies/mL)
Control mAb
ADG20 (25 mg/kg)
ADG20 (5 mg/kg)
<LOD (~10^5 copies/mL)
Time Post Exposure (Days)

LOD, limit of detection.
Dotted line represents LOD.

Figure 5. Impact of ADG20 on viral titre in Rhesus macaques

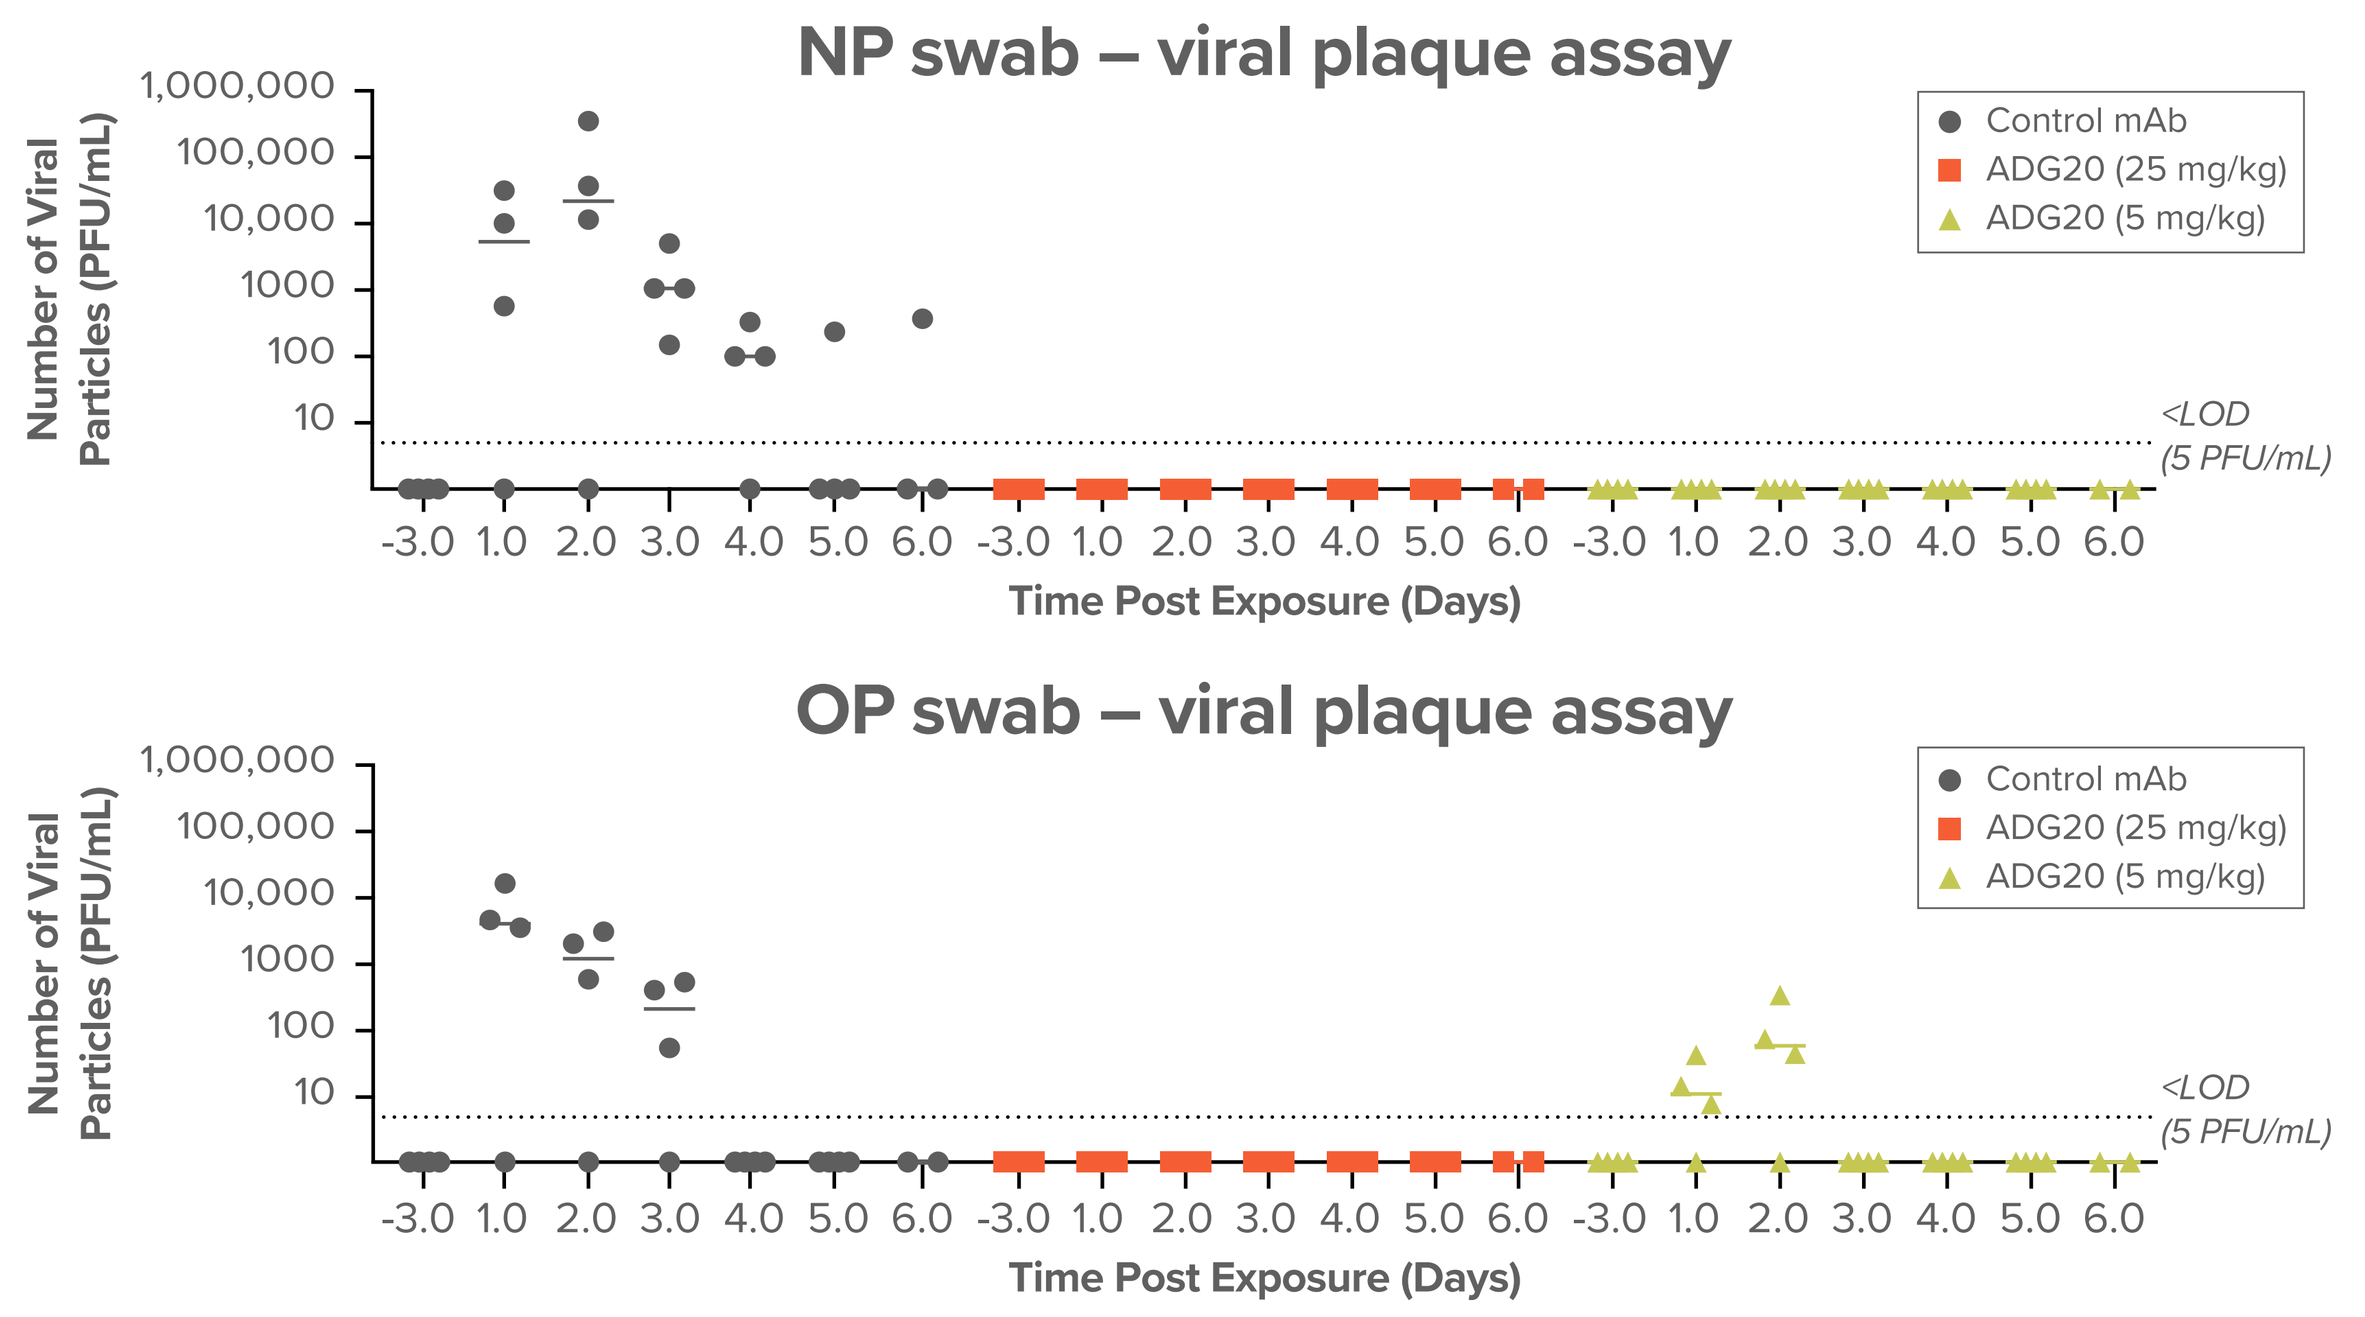
NP swab – viral plaque assay
OP swab – viral plaque assay
Number of Viral Particles (PFU/mL)
Control mAb
ADG20 (25 mg/kg)
ADG20 (5 mg/kg)
<LOD (5 PFU/mL)
Time Post Exposure (Days)

Dotted line represents LOD.

REFERENCES

1. Lauring AS, Hodcroft EB. *JAMA*. 2021;325:529-531.
2. Mortazavi A, et al. *Radiation*. 2021;1:18-32.
3. Rappazzo CT, et al. *Science*. 2021;371:eabf4830.
4. Dejnirattisai W, et al. *Cell*. 2021;S0092–8674(21)00428-1.

Acknowledgments

This study was funded by Adagio Therapeutics, Inc. Writing assistance was provided by Georgiana Manica, BSc, of Parexel, and was funded by Adagio Therapeutics, Inc.

DISCLOSURES

LMW is an inventor on a patent application submitted by Adagio Therapeutics, Inc., describing the engineered SARS-CoV-2 antibody. KN, CIK, LC, EH, and IY are employees of Adagio Therapeutics, Inc. ZM has received consulting fees from Adagio Therapeutics, Inc. ASH, EEZ, CMO, RRB, SEZ, and JMD have no conflicts to disclose. Opinions, interpretations, conclusions, and recommendations are those of the author and are not necessarily endorsed by the US Army.

KEY FINDINGS

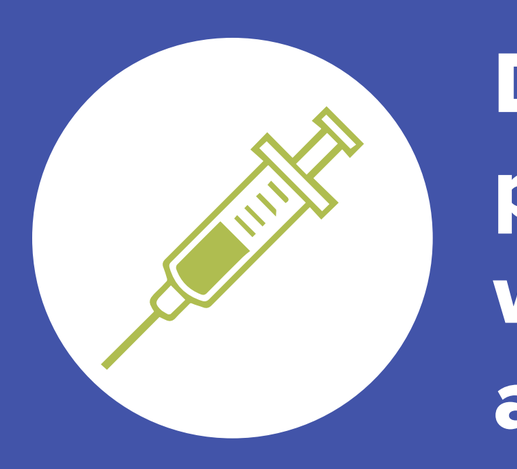

Dose-dependent prophylactic protection from SARS-CoV-2 infection was demonstrated in two different animal models

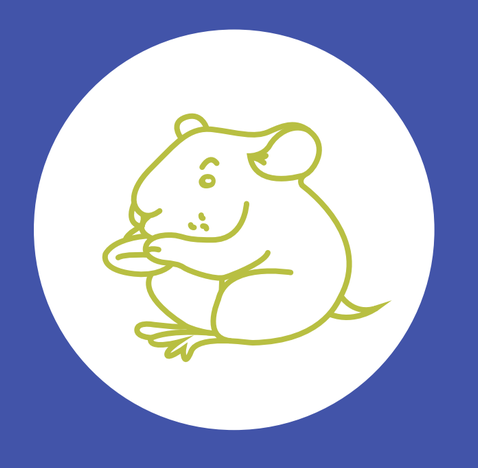

In the hamster model, ADG20 conferred significant protection from weight loss and lung pathology and inhibited viral replication at the 333 and 2000 ug doses versus control mAb

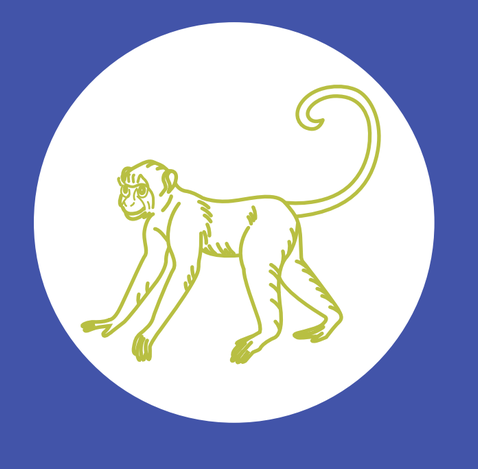

In the NHP model, viral replication was substantially reduced in the upper and lower airways of Rhesus macaques treated with ADG20 at doses of 5 or 25 mg/kg versus control mAb

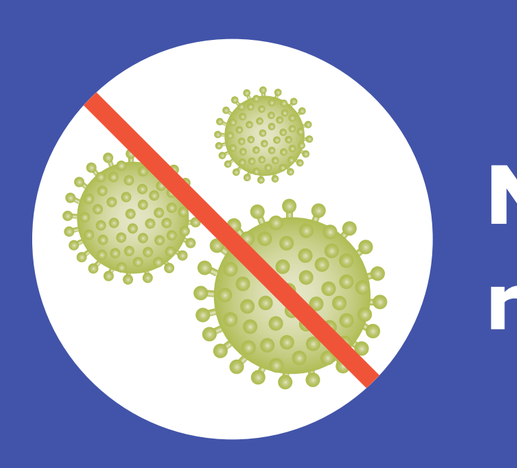

No evidence of enhanced viral replication or disease pathology

Scan the QR code to download an electronic version of the poster and to access supplemental content. The QR code is intended to provide scientific information for individual reference. The PDF should not be altered or reproduced in any way.

CONCLUSIONS

- Prophylactic administration of ADG20 provides potent protection against SARS-CoV-2 infection in two animal models
- The results support further investigation of ADG20 for the prevention of COVID-19 in humans
- Ongoing clinical trials seek to confirm the safety and efficacy of ADG20 in both the prevention (EVADE, ClinicalTrials.gov Identifier: NCT04859517) and treatment (STAMP, ClinicalTrials.gov Identifier: NCT04805671) of COVID-19

Poster presented at the virtual 31st European Congress of Clinical Microbiology & Infectious Diseases (ECCMID); July 9-12, 2021